CDK9 inhibition by KB-0742 is selective for transcriptionally addicted tumors harboring *MYC* genomic amplifications

Melinda A. L. Day[1], Nikolaus D. Obholzer[1], Akanksha Pandey[1], Tom Chen[1], Tong Liang[1], Rosa A. Villagomez[1], Christina S. Lee[1], Mulini Pingili[1], Jost V. Koren[2], David B. Freeman[1], Holly M. Nguyen[3], Jennifer L. Connor[3], Eva Corey[3], Matthew G. Reese[4], Andrew Boghossian[4], Brienne Engel[4], Melissa M. Ronan[4], Jennifer A. Roth[4], Joesph P. Vacca[1], Peter B. Rahl[1], Marius S. Pop[1], Benjamin W. Trotter[1], Charles Y. Lin[1], Jorge C. DiMartino[1], Pavan Kumar[1], Douglas C. Saffran[1].

[1]Kronos Bio, Cambridge, MA, [2]Baylor College of Medicine, Houston, TX, [3]University of Washington, Seattle, WA, [4]Broad Institute, Cambridge, MA

Abstract

Transcriptional deregulation is a hallmark of many cancers, including a subset that are “transcriptionally addicted” and depend on high levels of transcription for oncogenic program genes. To better define molecular sensitivity to transcriptional inhibition, we profiled pan-cancer sensitivity to KB-0742 — a potent, selective, and orally bioavailable small molecule inhibitor of the transcription elongation cofactor CDK9. Sensitivity profiling across ~800 adherent and suspension immortalized cell lines using the Broad PRISM platform revealed *MYC* genomic amplification as a key driver of CDK9 inhibitor sensitivity. MYC associated sensitivity was further observed in *ex vivo* primary patient tumor cell cultures and patient derived xenografts regardless of pre-treatment status. Analysis of the temporal kinetics of CDK9 inhibition revealed a rapid collapse of oncogenic transcription programs comprised largely of short half-life transcripts including key oncogenes such as *MYC* and *MCL1* and was followed by apoptosis at later time points. These data suggest that MYC genomic amplification may serve as an important feature defining sensitivity to CDK9 inhibition in patients with advanced solid tumors.

KB-0742 is a selective CDK9 inhibitor

A

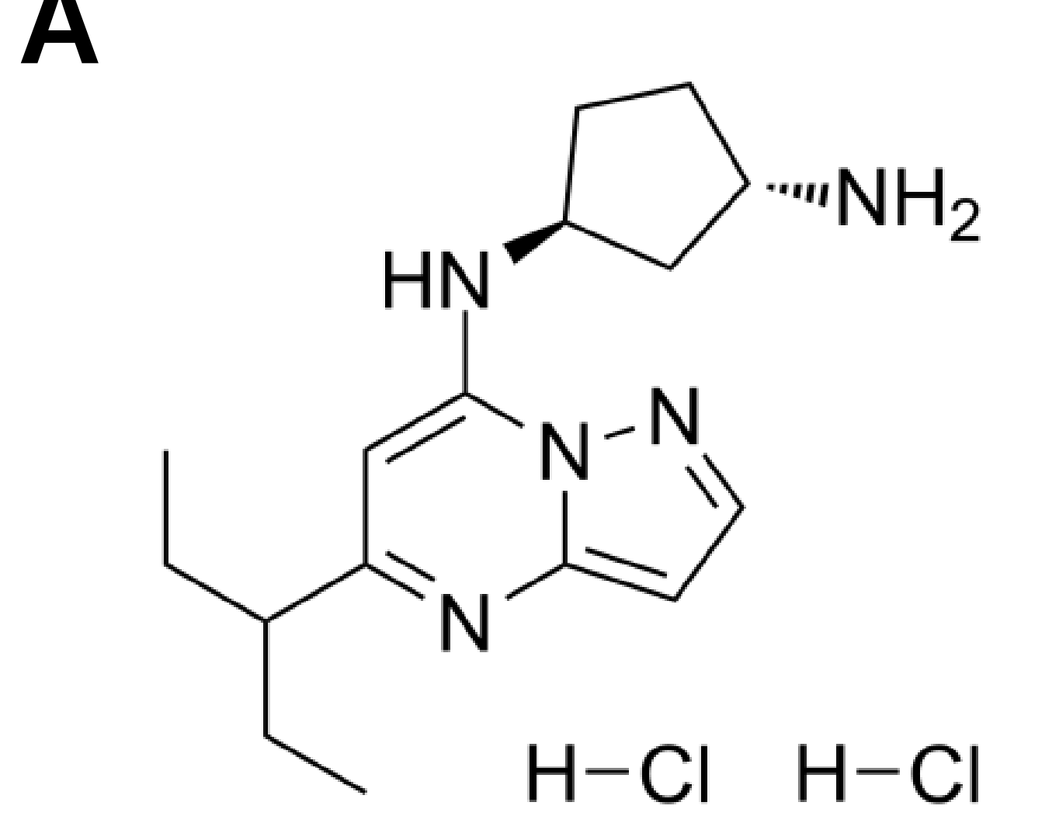
HN
NH2
N
N
N
H−Cl H−Cl

B

Kinase	KB-0742 Potency (nM)
CDK9	6
CDK8	6,000
CDK7	1,512
CDK6	3,948
CDK5	1,818
CDK4	3,132
CDK3	1,422
CDK2	396
CDK3	2,982

Figure 1: KB-0742 is a selective inhibitor of CDK9. (A) Structure of KB-0742. (B) Heat map of potency (biochemical IC50) of KB-0742 against different cyclin dependent kinases. KB-0742 is selectively active against CDK9 with a single digit nanomolar IC50. KB-0742 was tested against an additional panel of 631 other kinases with all showing high IC 50 values.

CDK9 – MYC Translational Network

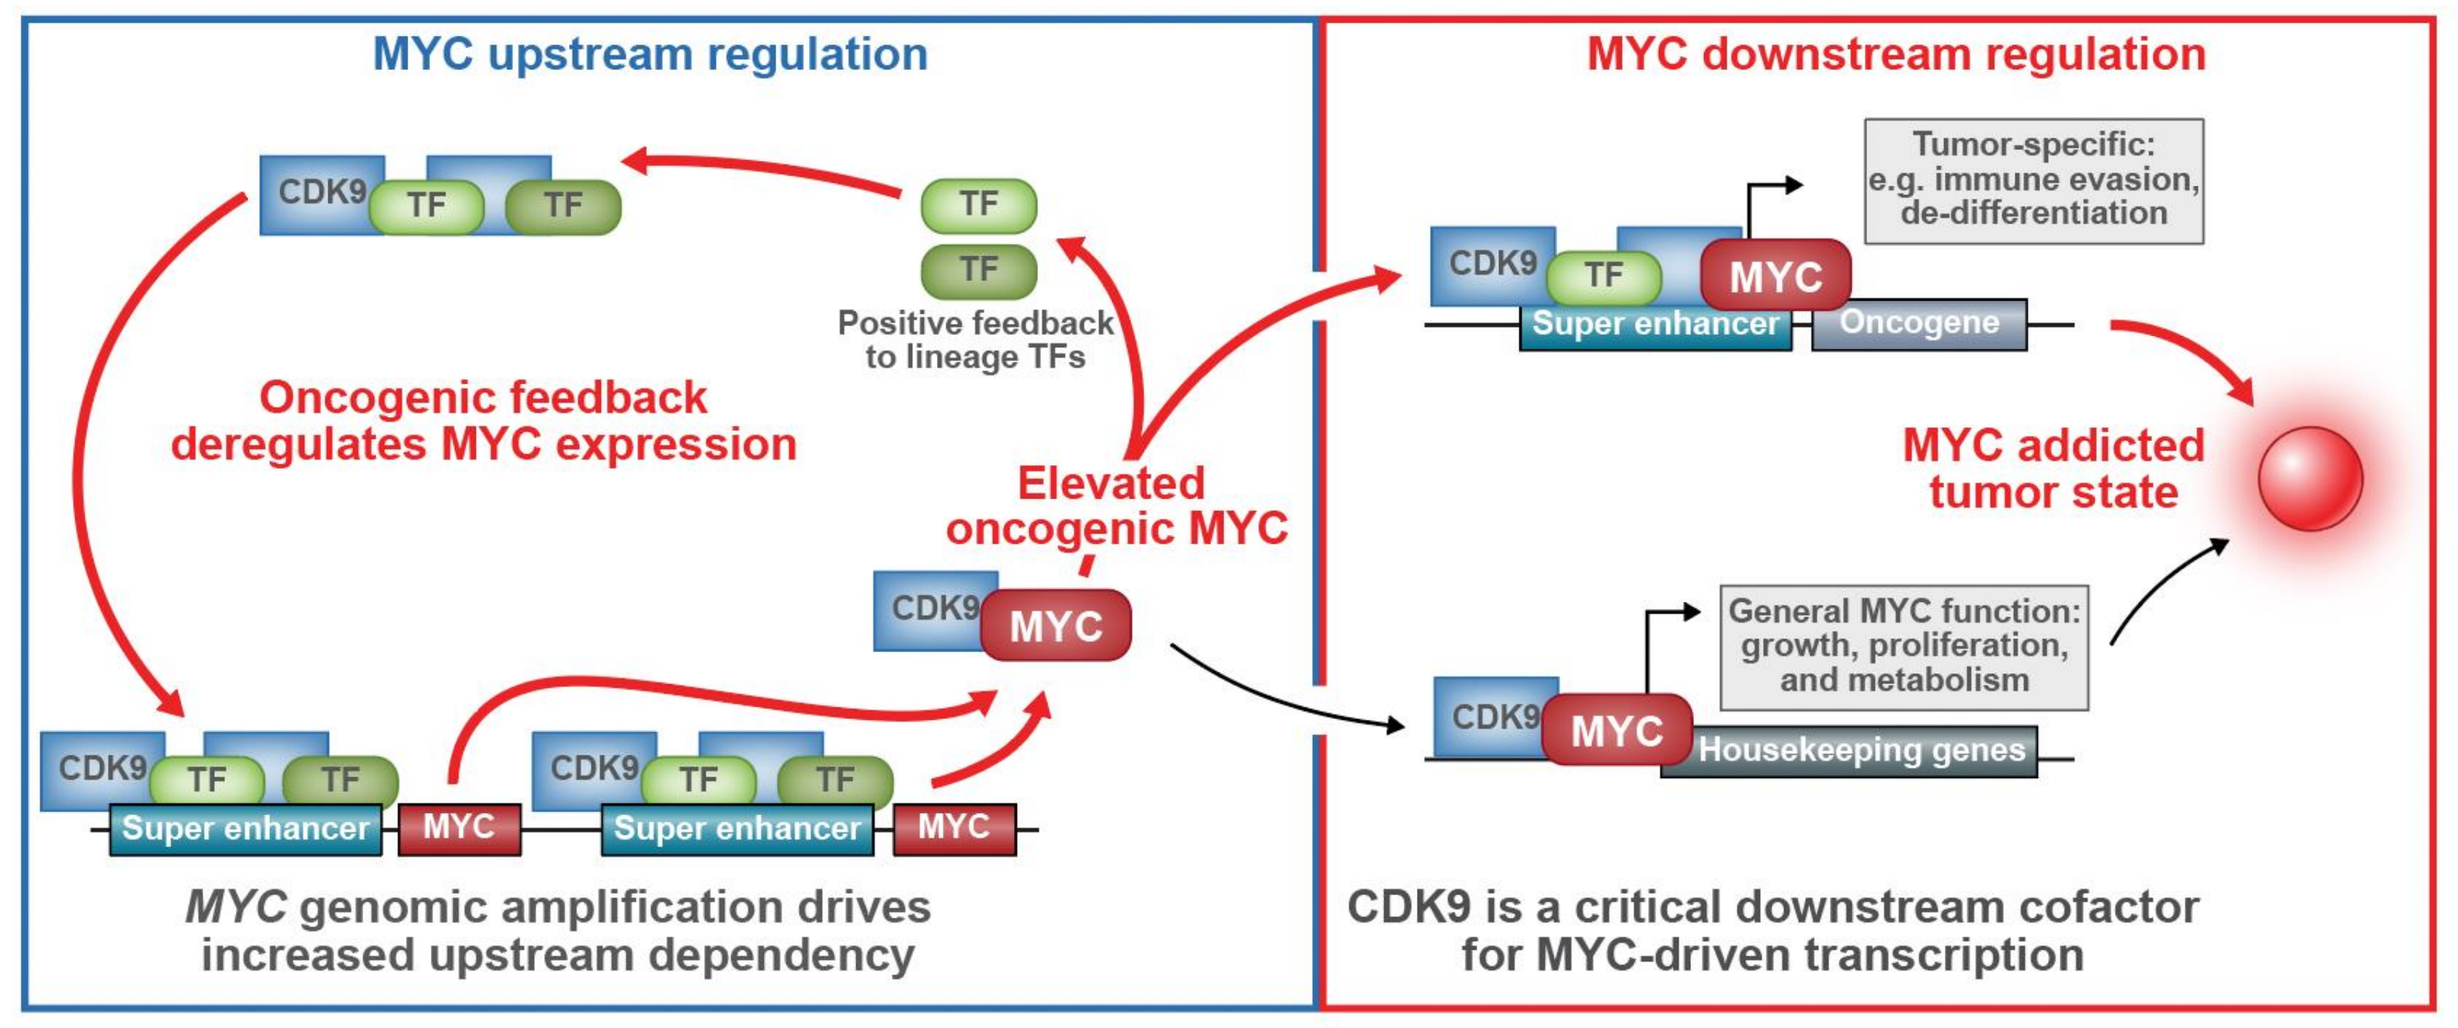
MYC upstream regulation
MYC downstream regulation
Oncogenic feedback deregulates MYC expression
Positive feedback to lineage TFs
Elevated oncogenic MYC
MYC genomic amplification drives increased upstream dependency
Tumor-specific: e.g. immune evasion, de-differentiation
MYC addicted tumor state
General MYC function: growth, proliferation, and metabolism
Super enhancer
Oncogene
Housekeeping genes
CDK9 is a critical downstream cofactor for MYC-driven transcription

Figure 2: CDK9 is both an upstream and downstream cofactor of MYC in *MYC* amplified tumors. Left: lineage-specific transcription factors recruit CDK9 to super enhancers at the *MYC* amplicon, driving high levels of MYC expression. Center: elevated oncogenic MYC activates lineage-specific transcription factors, creating a positive feedback loop. Right: CDK9 is an essential cofactor of MYC and allows MYC to drive both tumor-specific and normal gene expression programs.

KB-0742 is active in MYC amplified cell lines

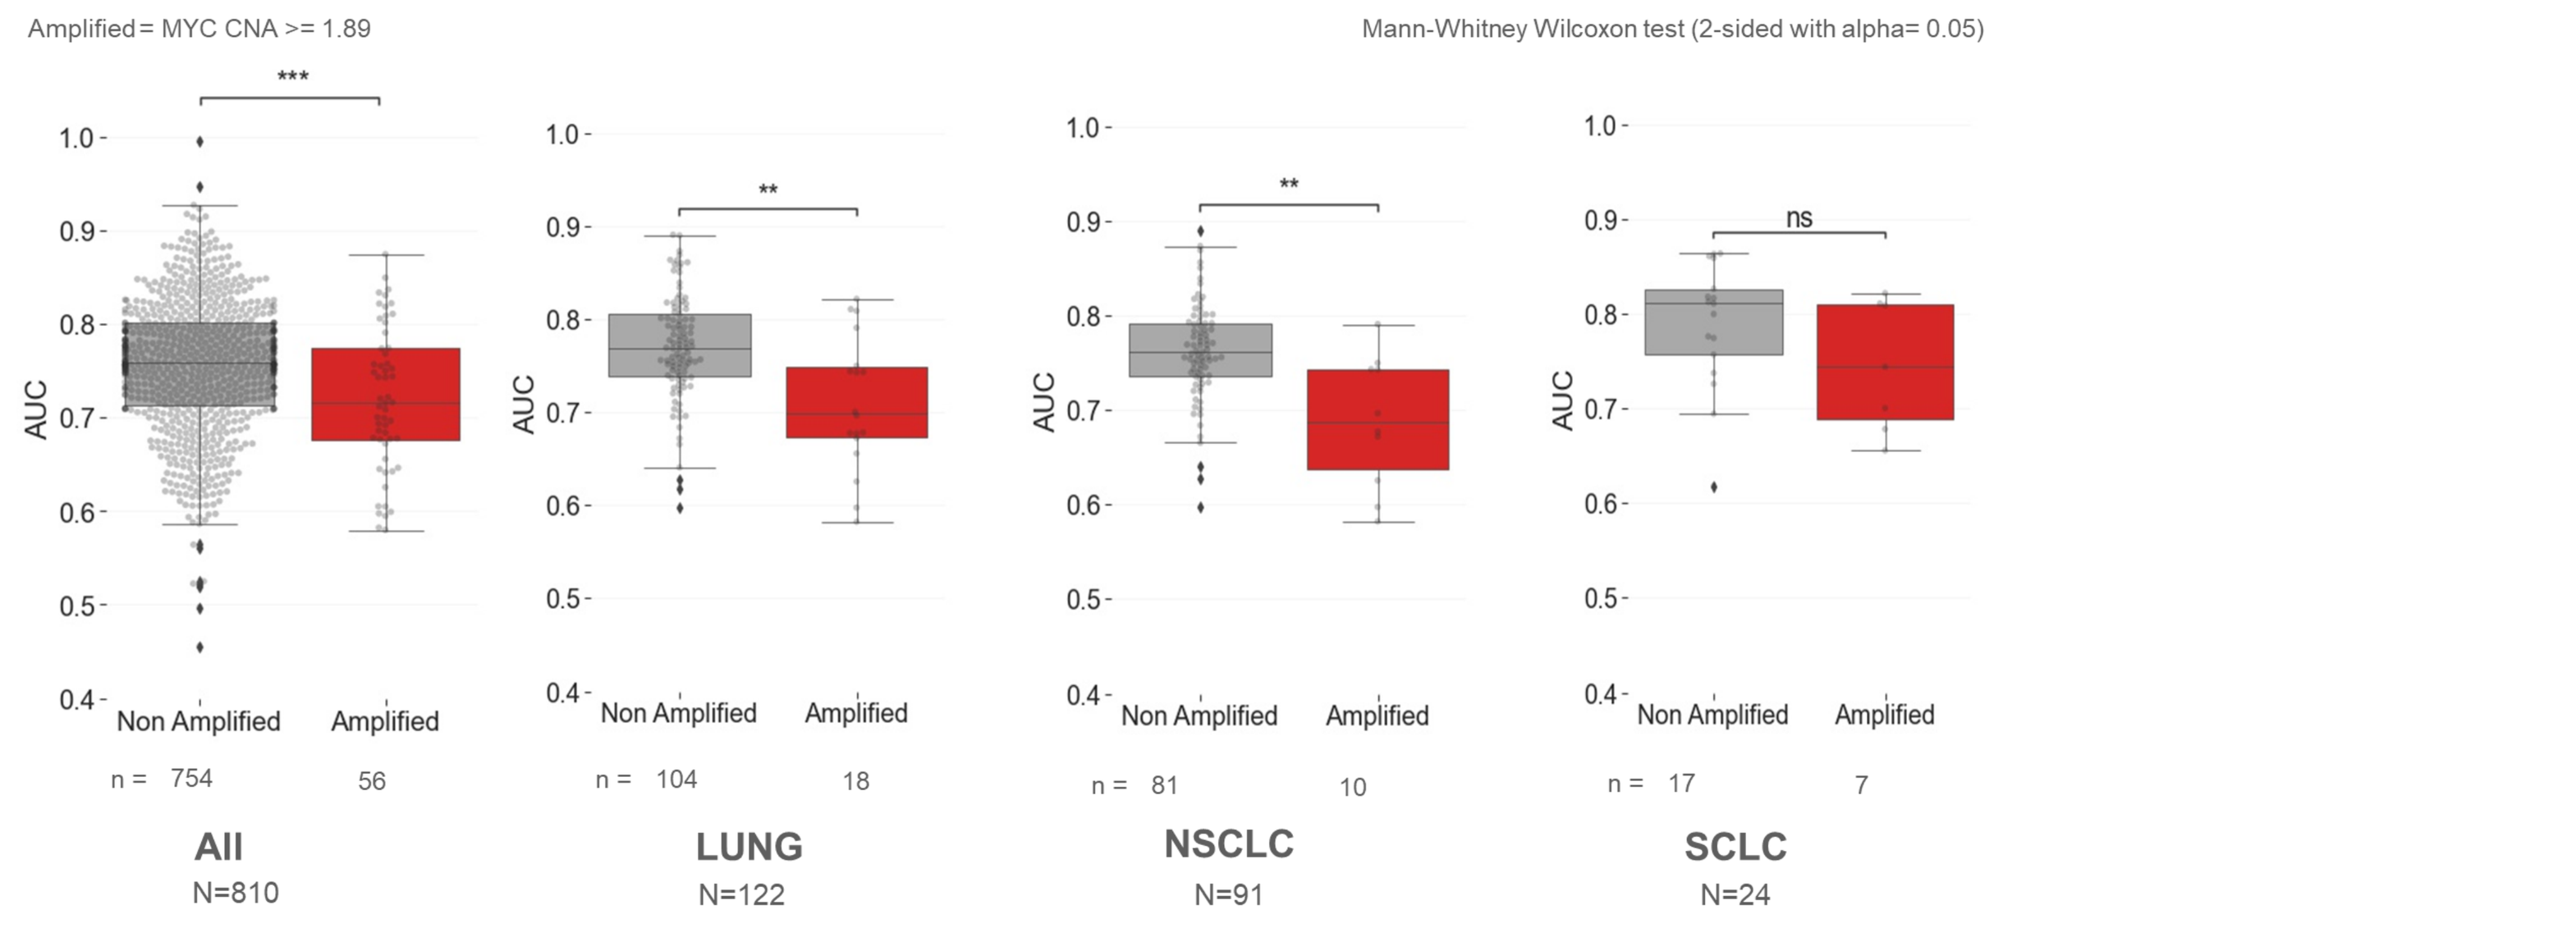
Amplified= MYC CNA >= 1.89
Mann-Whitney Wilcoxon test (2-sided with alpha= 0.05)
AUC
Non Amplified
Amplified
n = 754 56
n = 104 18
n = 81 10
n = 17 7
All N=810
LUNG N=122
NSCLC N=91
SCLC N=24

Figure 3: *MYC* amplified lines have a lower AUC compared to non-amplified cell lines. The figure shows boxplots for *MYC* CNA categories with AUC values for all lineages, lung, NSCLC and SCLC for KB-0742. Red color indicates *MYC* amplified, and grey indicates *MYC* non-amplified categories. Two-sided Mann Whitney Wilcoxon test with alpha= 0.05 was performed between amplified and non-amplified categories.

Pan cancer activity of KB-0742 observed in PDCs

Cancer Type	# of Models	Treatment History: # Models per Lines of Previous Therapy				
		Naive	1 line	2 lines	3+ lines	Unknown
Breast	23	4	5	2	10	2
Ovarian	19	5	1	5	8	
Colon	9	2	2	2	2	1
Prostate	17	16	1			
Melanoma	4	1	1		1	1
Liver	1			1		
Sarcoma	4	1		1		2
Glioblastoma (GBM)	3	3				
Pancreatic	3		2		1	
Lung	15	12	1		2	
Rectal	1				1	
Endometrial	1			1		
Renal Cell Carcinoma (RCC)	1	1				
Neuroendocrine	1				1	

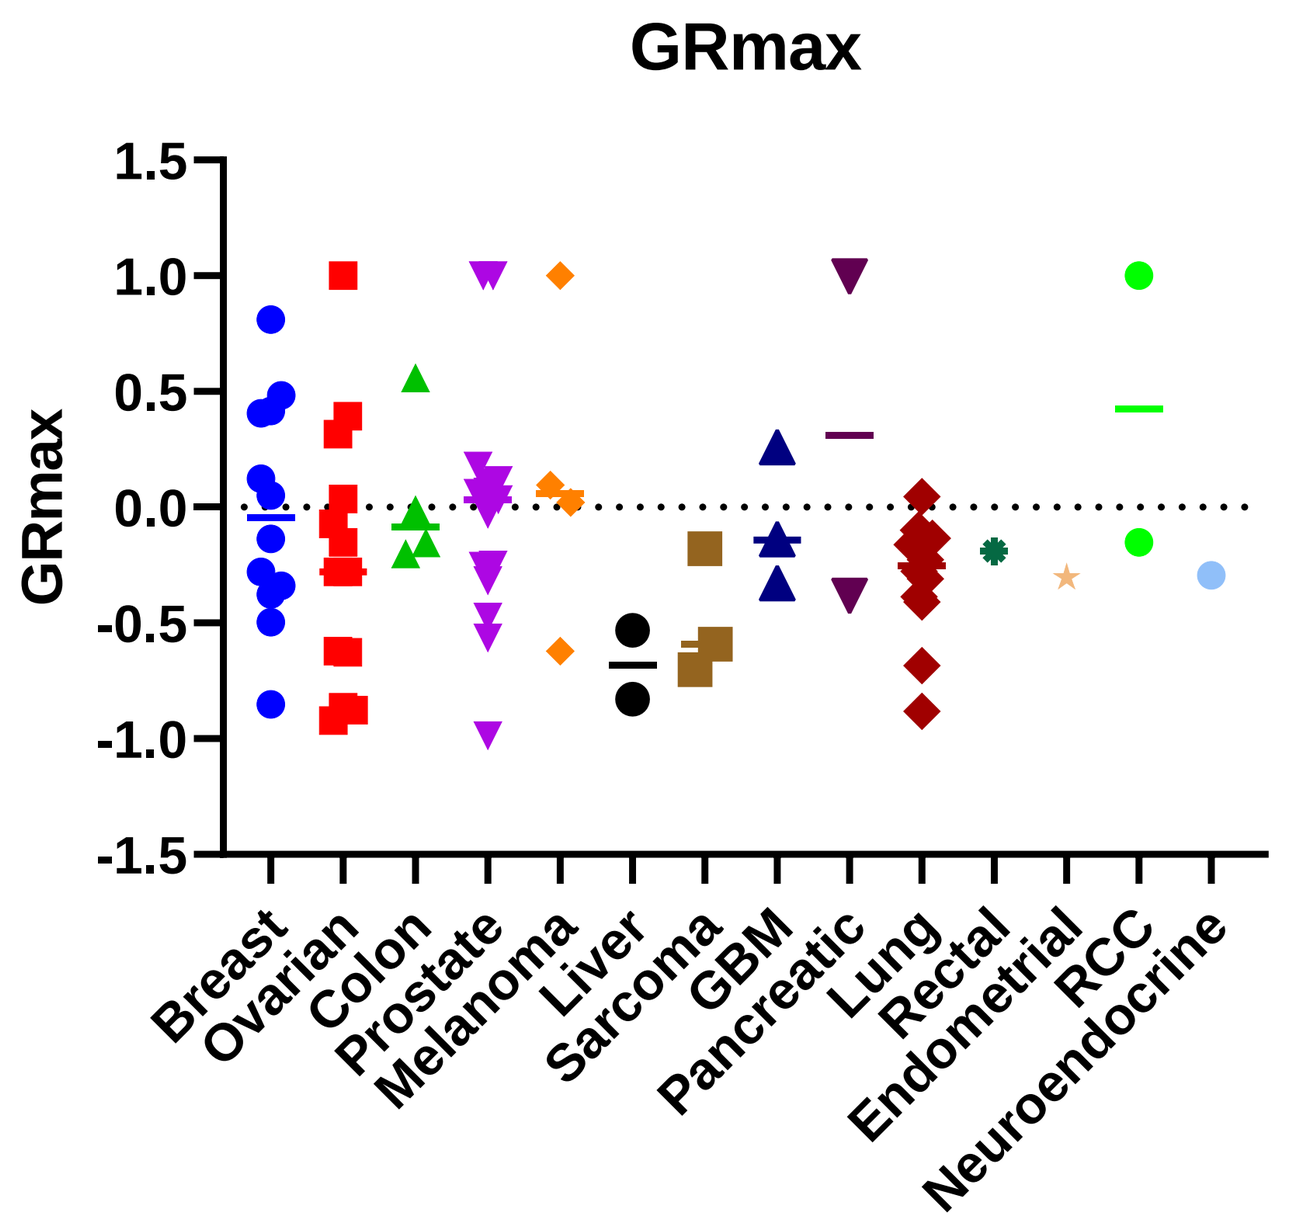
GRmax
GRmax
Breast
Ovarian
Colon
Prostate
Melanoma
Liver
Sarcoma
GBM
Pancreatic
Lung
Rectal
Endometrial
RCC
Neuroendocrine

Figure 4: KB-0742 shows pan cancer activity across a variety of patient derived cell lines (PDCs) with various treatment histories. 100 PDC models with different treatment histories (table) and MYC expression levels were tested for sensitivity to KB-0742. Each model was treated with KB-0742 titration curve and analyzed for cell number, cell death, and cell growth. Growth rate inhibition analysis showed KB-0742 induced cytotoxicity across several tumor types as measured by GRmax (values <0 indicates cytotoxicity).

KB-0742 is active in MYC expressing PDOs

A

Model Number	Indication	Treatment history	MYC TPM
KOLU-045	Small Cell Lung Cancer	Naïve	70
KOLU-299		Naïve	30
KOLU-448		Lobaplatin+Etoposide	30
KOLU-775H		Cisplatin	20
KOLU-545H		VP16+Lobaplatin	68
KOLU-643H		VP16 + Lobaplatin	88
KOBR-011	Triple Negative Breast Cancer	TNBC: EPI + PTX 6 cycle	UNK
KOBR-472		TNBC: PTX + CBP 4 cycle	UNK

Maximum % Inhibition

	Cisplatin	Pemetrexed	Paclitaxel	Gemcitabine	KB-0742
KOLU-045	10.52	12.83	44.81	53.02	99.99
KOLU-299	10.00	10.00	48.42	57.21	94.19
KOLU-448	10.00	18.97	21.28	34.95	99.02
KOLU-775H	10.00	10.00	49.61	71.74	94.69
KOLU-545H	11.57	4.79	17.50	25.06	95.88
KOLU-643H			16.29	No effect	70.65
KOBR-011	No effect		31.56	59.99	100.00
KOBR-472			No effect	15.06	89.00

B

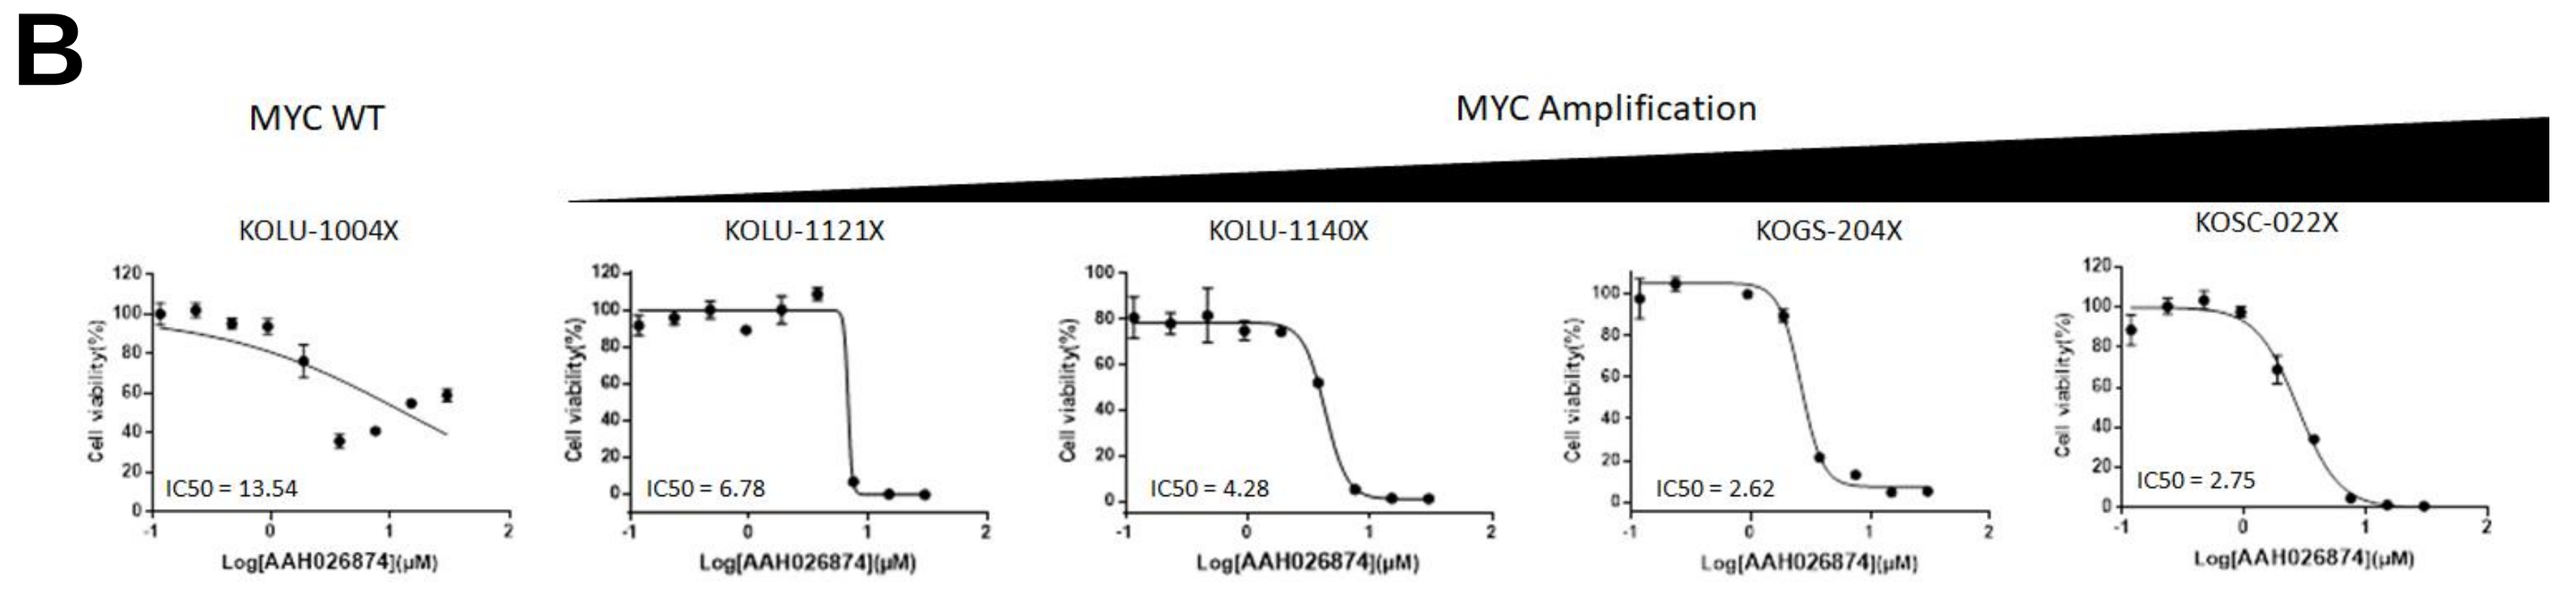
MYC WT
MYC Amplification
KOLU-1004X
KOLU-1121X
KOLU-1140X
KOGS-204X
KOSC-022X
IC50 = 13.54
IC50 = 6.78
IC50 = 4.28
IC50 = 2.62
IC50 = 2.75

C D

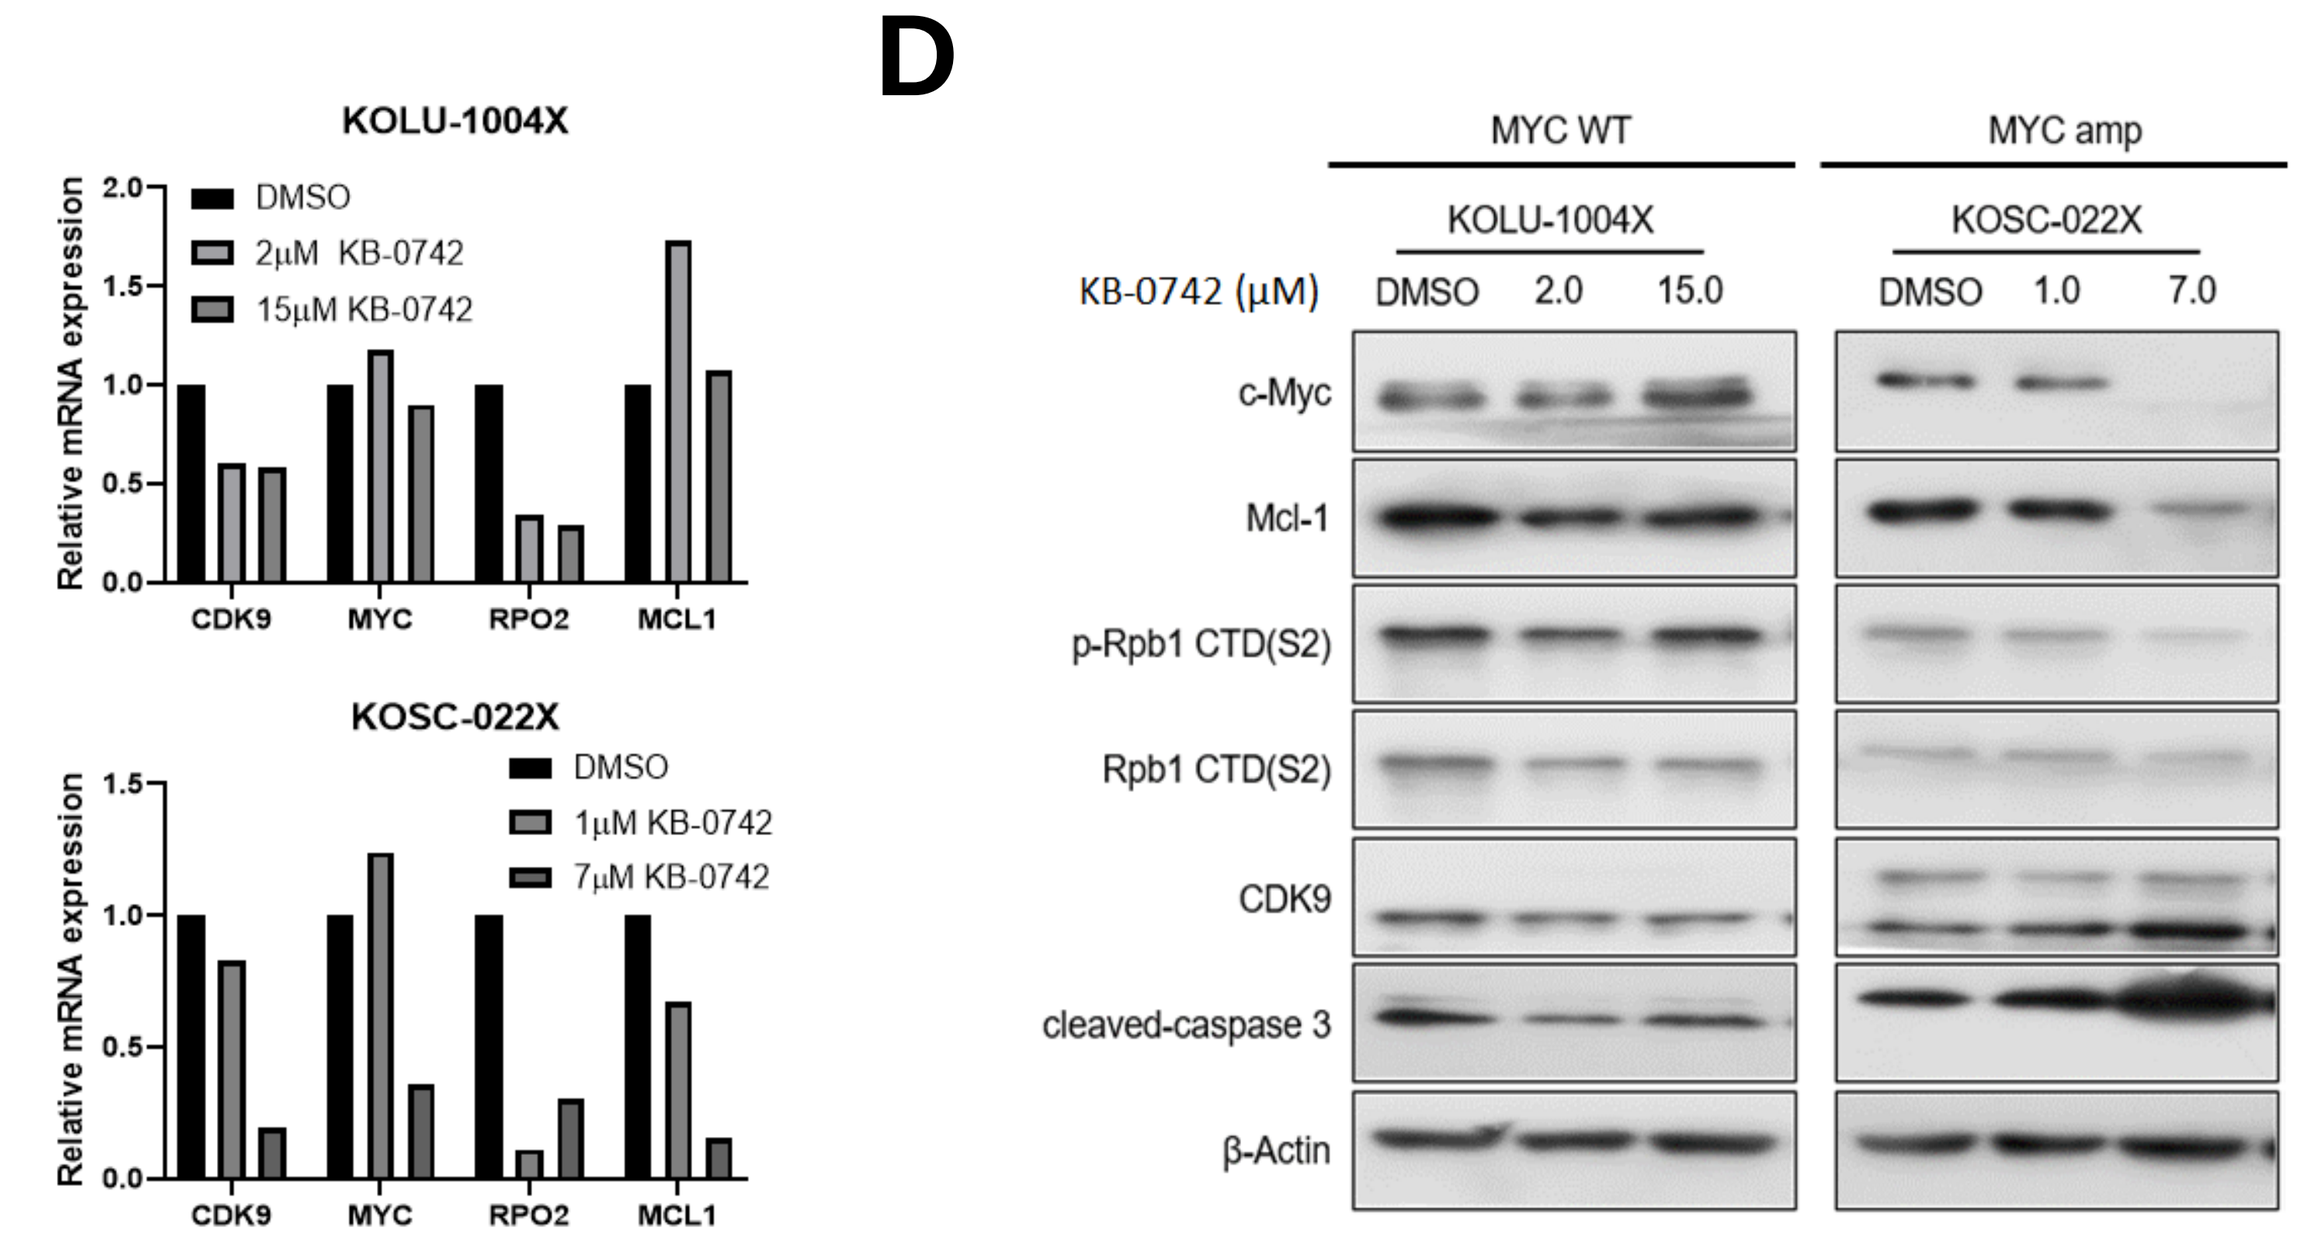
KOLU-1004X
DMSO
2μM KB-0742
15μM KB-0742
KOSC-022X
1μM KB-0742
7μM KB-0742
Relative mRNA expression
CDK9
MYC
RPO2
MCL1
MYC WT
MYC amp
KB-0742 (μM)
c-Myc
Mcl-1
p-Rpb1 CTD(S2)
Rpb1 CTD(S2)
cleaved-caspase 3
β-Actin

Figure 5: KB-0742 is active in MYC expressing patient derived organoids (PDO). KB-0742 activity was measured in PDO models. (A) A panel of six SCLC PDO models with various treatment histories was used to compare the activity of KB-0742 to four standard of care (SOC) compounds. KB-0742 showed greater inhibition of the models than all four SOC compounds. (B) PDO models of NSCLC (KOLU-1004X, KOLU-1140X, KOLU-1121X), Rhabdomyosarcoma (KOSC-022X), and Gastric (KOGS-240X) cancers were treated with KB-0742 and the IC50 calculated. KB-0742 sensitivity correlated with MYC amplification. (C and D) KOLU-1004X and KOSC-022X were treated with KB-0742 at the IC50 and IC90 for each model for 8 hours. (C) KB-0742 resulted in a reduction in the gene expression of *CDK9* and *RPO2* in both models, and *MYC* and *MCL1* in the KOSC-022X. (D) Western blot showed reduction in c-MYC and MCL-1 protein in KOSC-022X , and an increase in cleaved caspase 3.

KB-0742 inhibits MYC expression profile *in vivo*

A

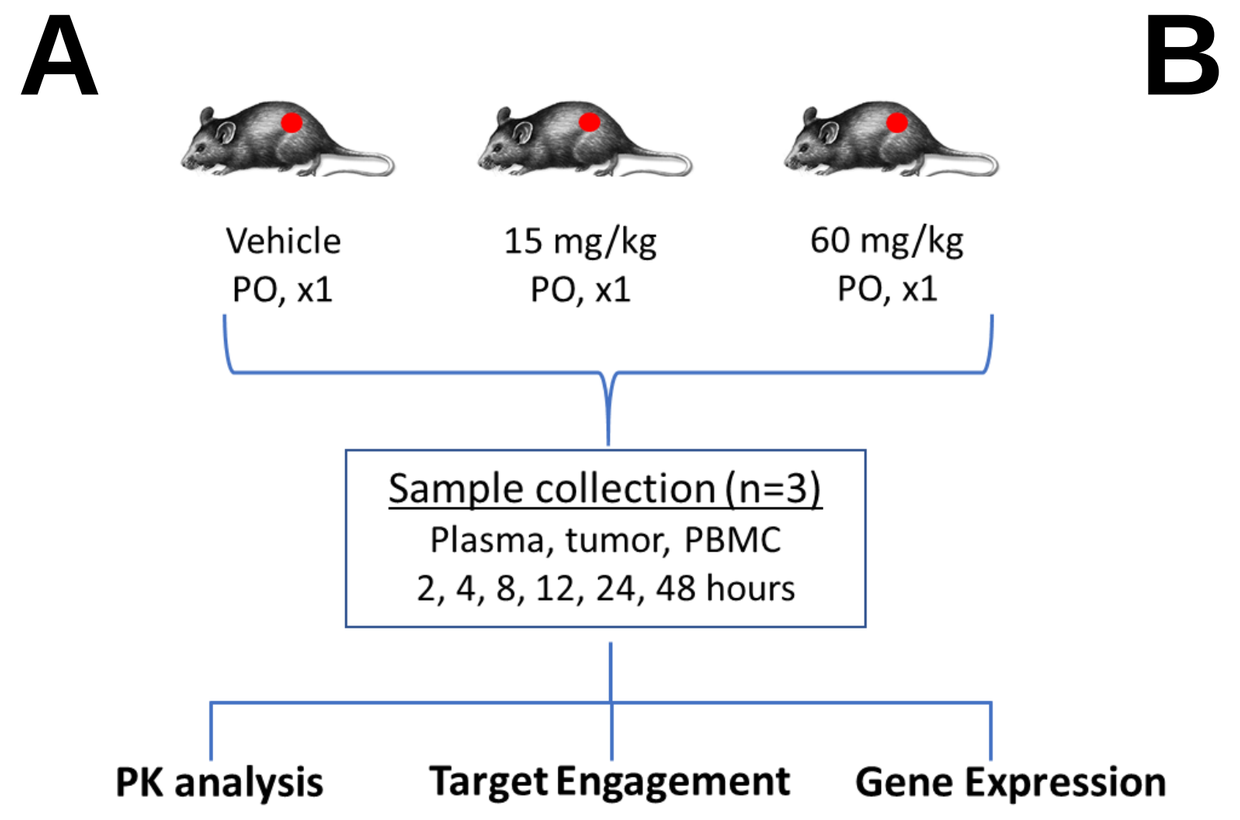
Vehicle PO, x1
15 mg/kg PO, x1
60 mg/kg PO, x1
Sample collection (n=3)
Plasma, tumor, PBMC
2, 4, 8, 12, 24, 48 hours
PK analysis
Target Engagement
Gene Expression

B C

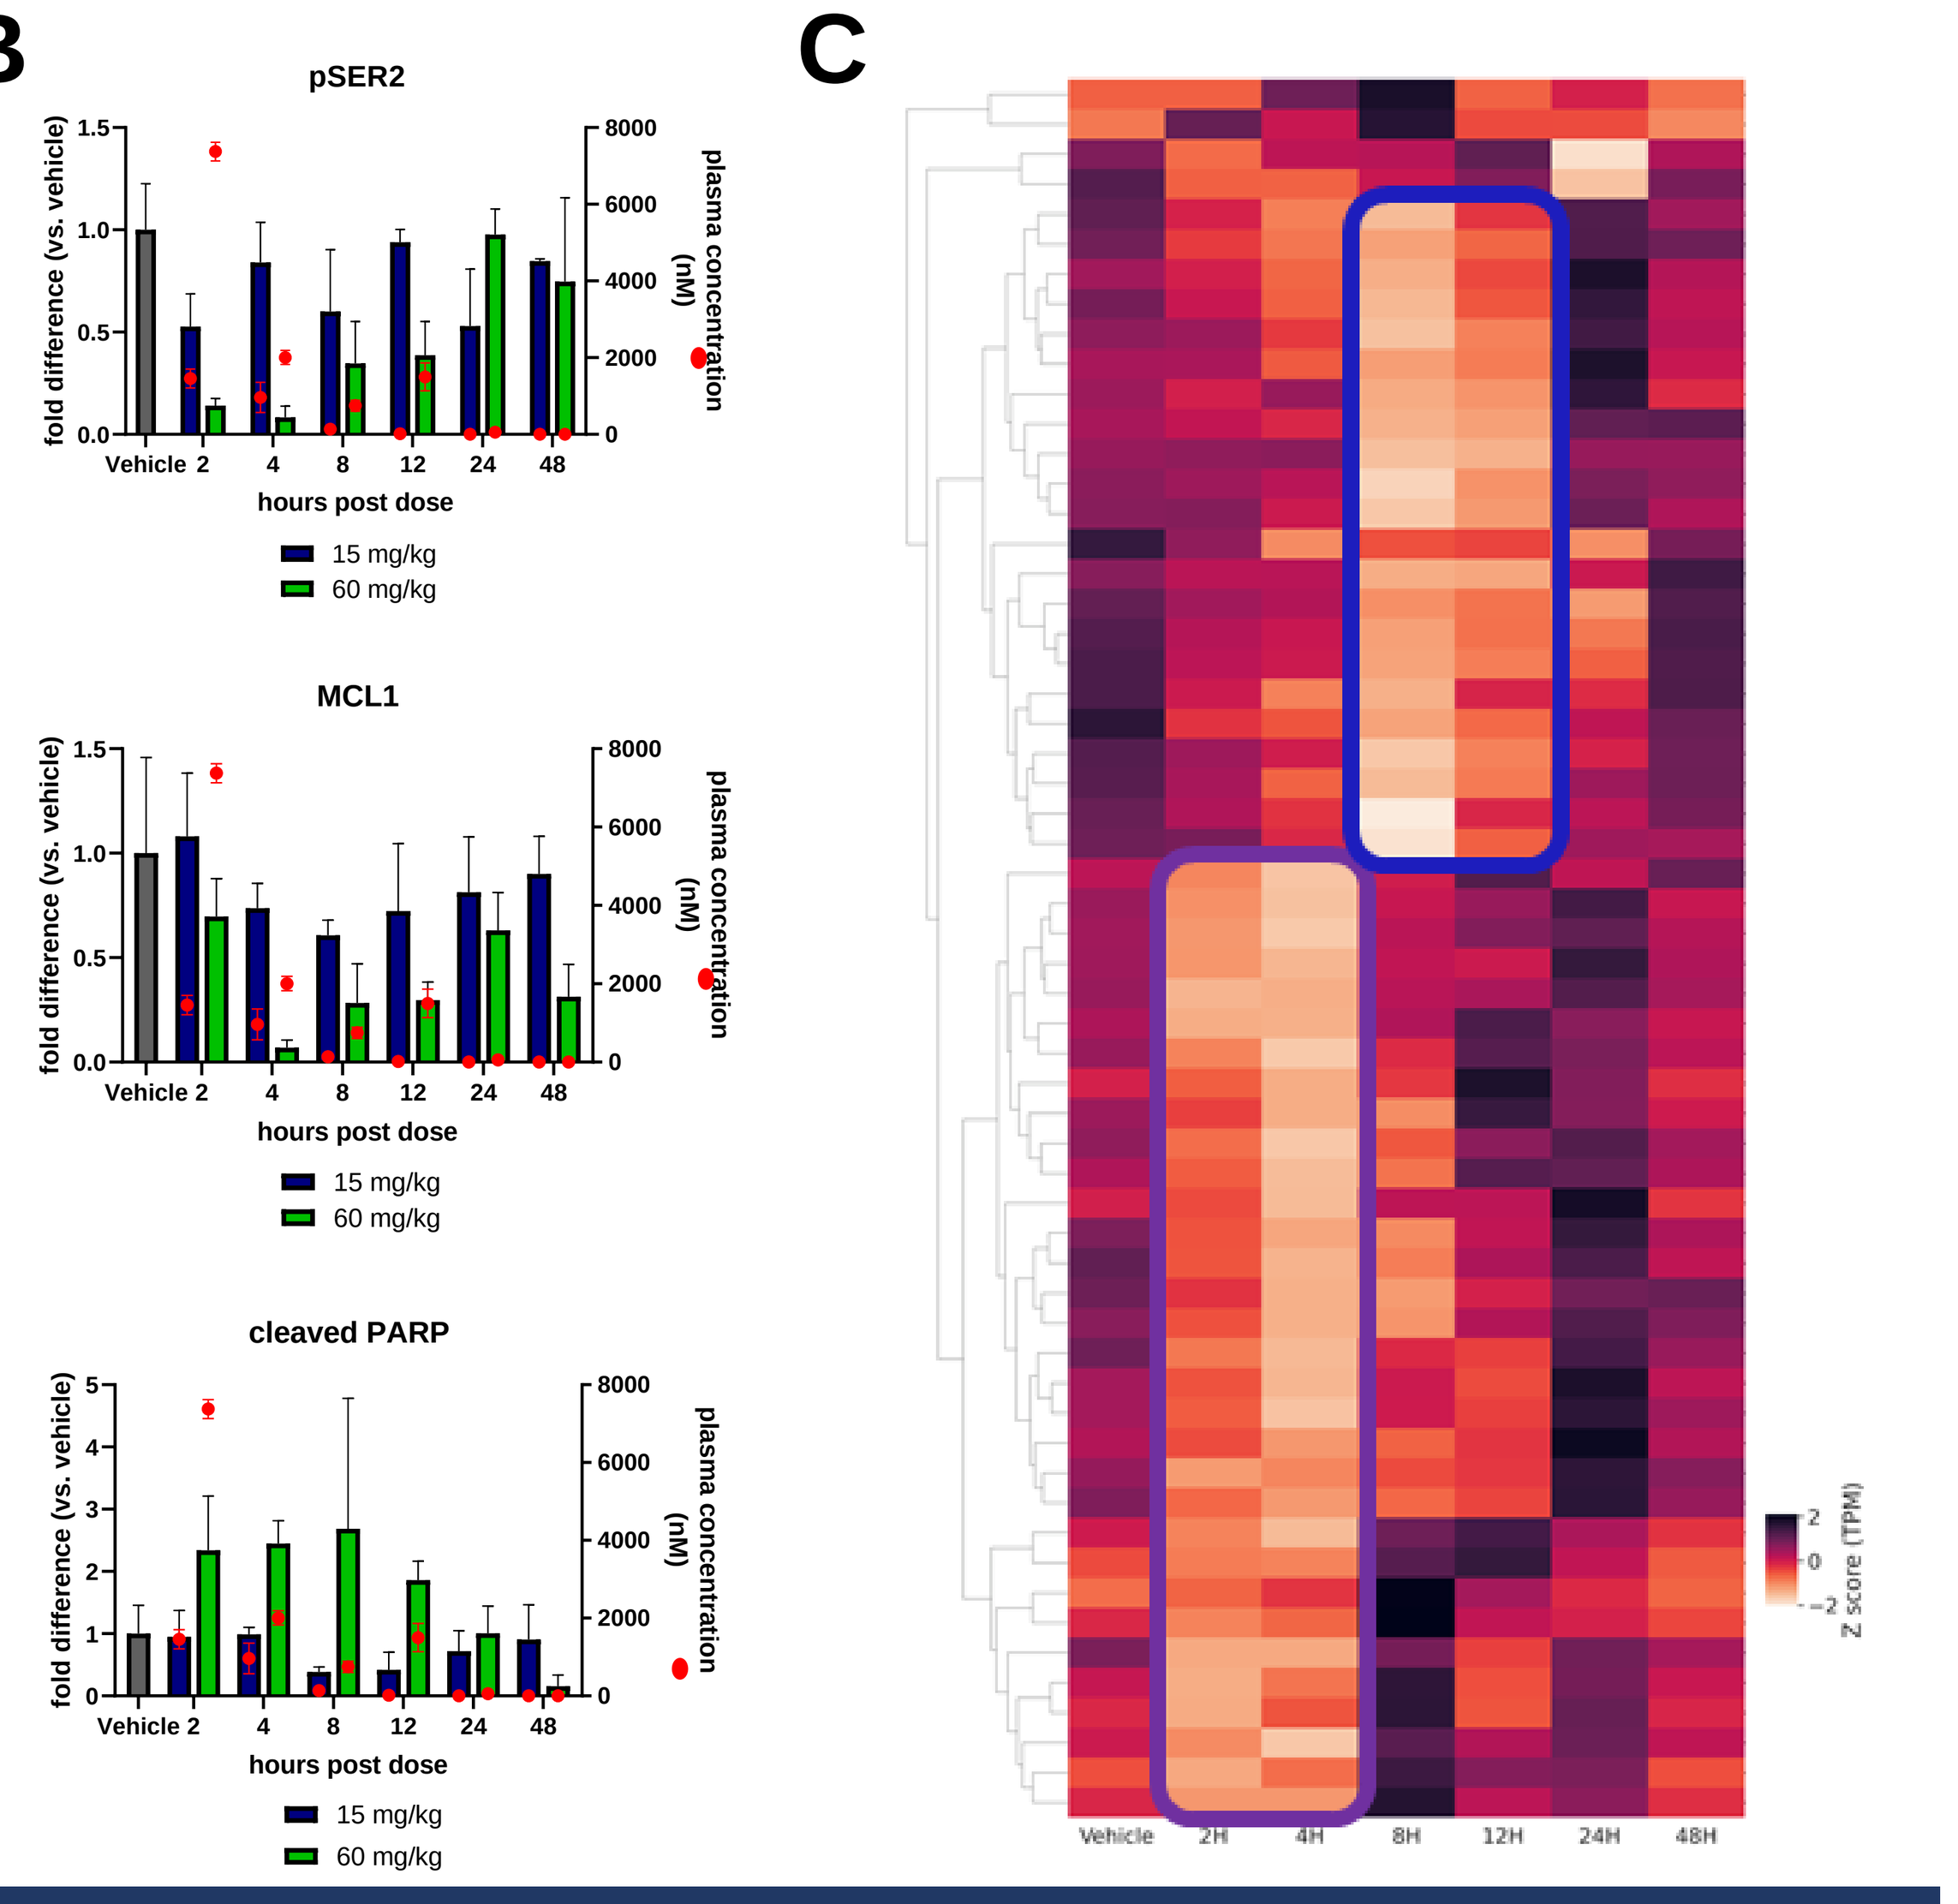
pSER2
MCL1
cleaved PARP
fold difference (vs. vehicle)
plasma concentration (nM)
hours post dose
15 mg/kg
60 mg/kg
Vehicle
2H
4H
8H
12H
24H
48H
Z score (TPM)

Figure 6: KB-0742 inhibits CDK9 and reduces the MYC expression profile *in vivo*. (A) Mice with subcutaneous MV4-11 derived tumors were treated with a single indicated dose of KB-0742 and samples taken at the indicated time points. (B) KB-0742 treatment reduced pSER2 and MCL1 protein levels and increased cleaved PARP in a dose dependent manner. (C) KB-0742 treatment resulted in a clear reduction in expression of MYC targets genes; both early (2-4 hr; purple) and late (8-12 hr; blue) targets.

KB-0742 shows anti-tumor activity in PDX models

A

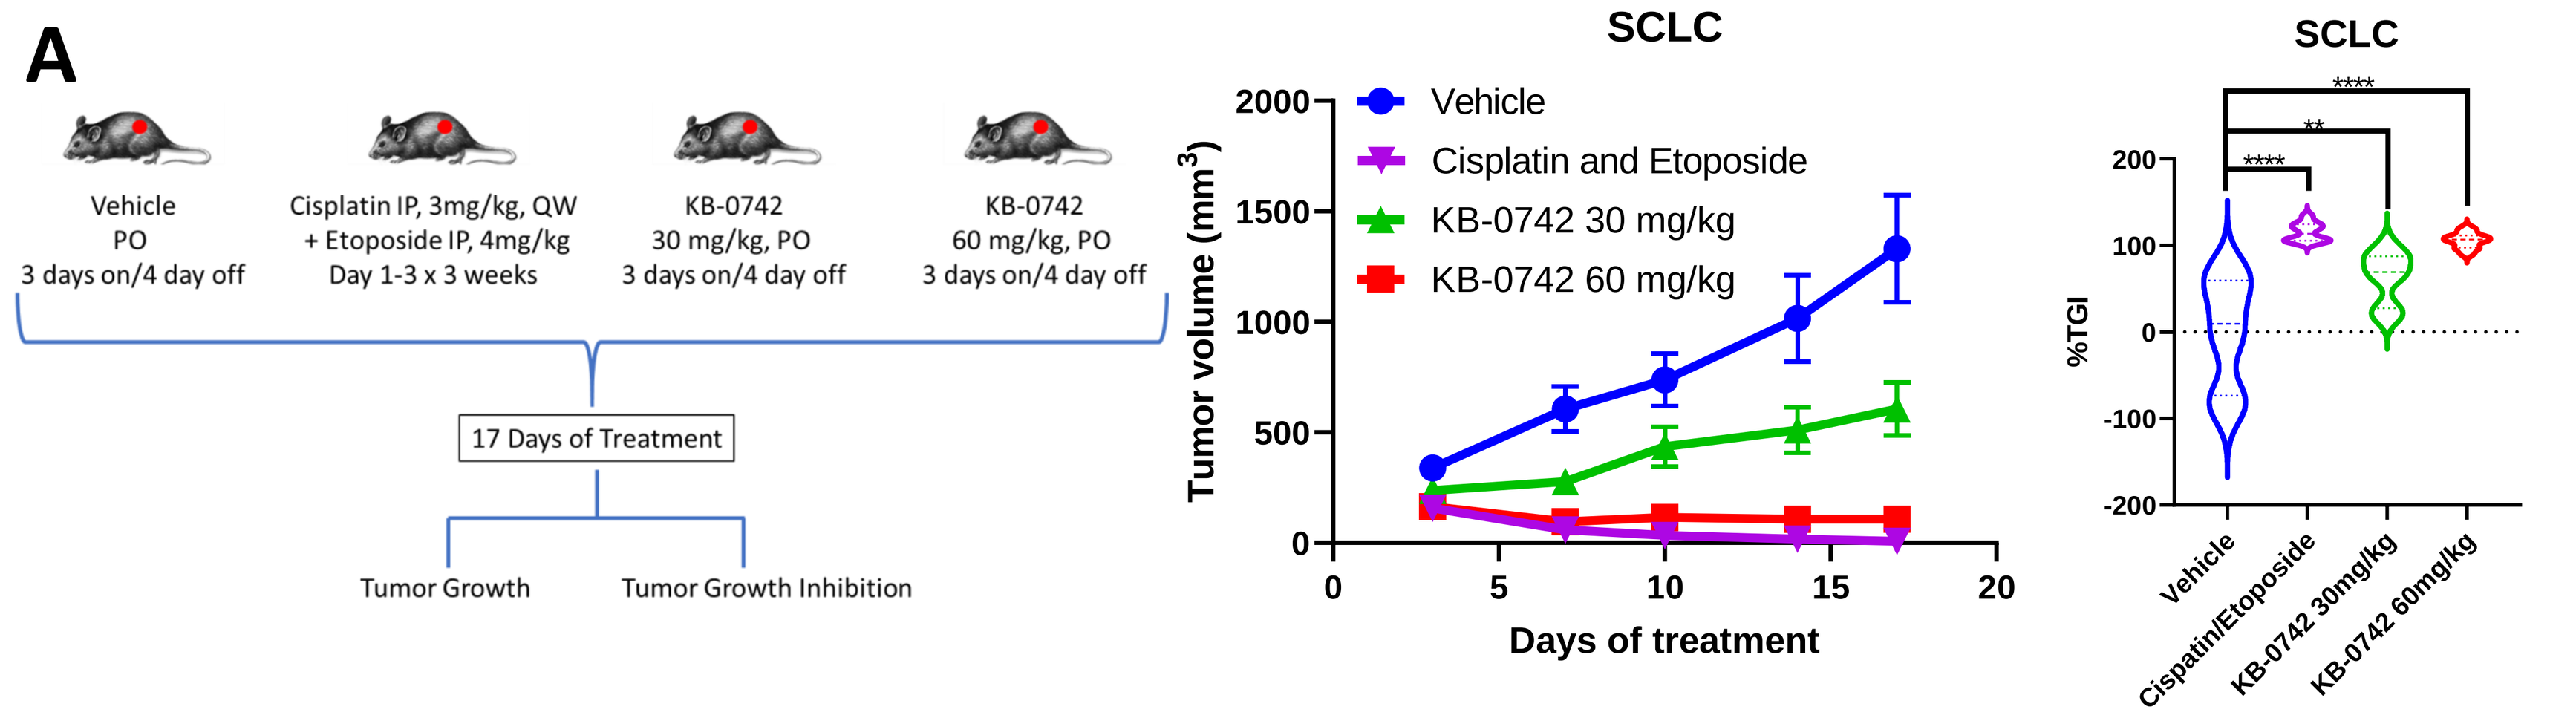
Vehicle PO 3 days on/4 day off
Cisplatin IP, 3mg/kg, QW + Etoposide IP, 4mg/kg Day 1-3 x 3 weeks
KB-0742 30 mg/kg, PO 3 days on/4 day off
KB-0742 60 mg/kg, PO 3 days on/4 day off
17 Days of Treatment
Tumor Growth
Tumor Growth Inhibition
SCLC
Vehicle
Cisplatin and Etoposide
KB-0742 30 mg/kg
KB-0742 60 mg/kg
Tumor volume (mm3)
Days of treatment
%TGI
Cispatin/Etoposide
KB-0742 30mg/kg
KB-0742 60mg/kg

B

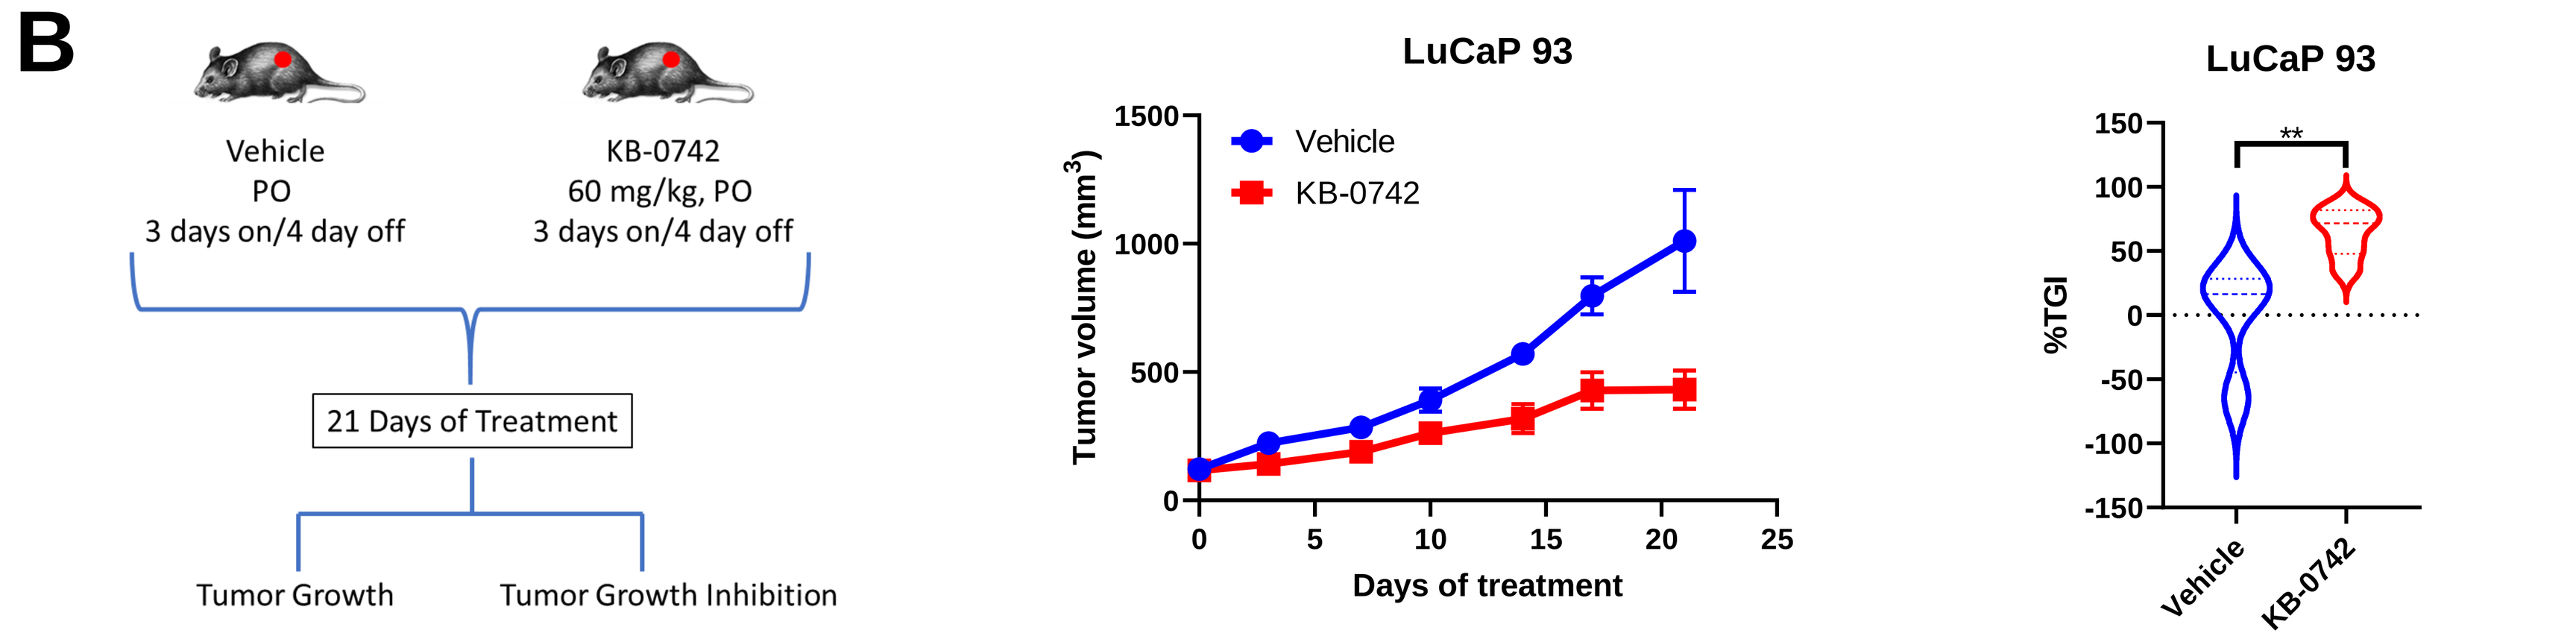
Vehicle PO 3 days on/4 day off
KB-0742 60 mg/kg, PO 3 days on/4 day off
21 Days of Treatment
Tumor Growth
Tumor Growth Inhibition
LuCaP 93
Vehicle
KB-0742
Tumor volume (mm3)
Days of treatment
%TGI

Figure 7: KB-0742 showed anti-tumor activity with intermittent dosing in MYC high expressing models. (A) Mice were subcutaneously engrafted with a patient derived tumor model of small cell lung cancer. Animals were treated with vehicle, cisplatin and etoposide, and KB-0742 at either 30 or 60 mg/kg. KB-0742 was given on an intermittent dosing schedule of 3 days on, 4 days off. Treatment with KB-0742 at 60 mg/kg showed similar anti-tumor activity to cisplatin and etoposide treatment with a tumor growth inhibition (TGI) of 105.7%. (B) Mice were subcutaneously engrafted with a model of neuroendocrine prostate cancer LuCaP 93 and treated with either vehicle or KB-0742 at 60 mg/kg on a 3 day on, 4 day off schedule. KB-0742 resulted in a TGI of 64.6%. Error bars represent standard error of the mean. **p<0.01,****p<0.0001

Conclusions

- KB-0742 is a selective CDK9 inhibitor and is active in models of MYC high expressing pan-cancers that have been exposed to multiple lines of therapy.
- KB-0742 treatment results in a reduction of *CDK9*, *pSER2*, *MYC* expression and inhibits the MYC expression profile both *in vitro* and *in vivo*.
- KB-0742 shows similar or greater levels of activity in SCLC models than SOC compounds both *ex vivo* and *in vivo*.
- KB-0742 intermittent dosing results in tumor growth inhibition in multiple MYC high expressing patient derived xenograft models.

A Phase 1/2 clinical trial of KB-0742 (NCT04718675) is currently recruiting patients with relapsed or refractory solid tumors or non-Hodgkin lymphoma